W3112

Electrospinning Optimisation of Drug-Loaded Eudragit E PO for Paediatric Oral Drug Delivery

H. Abdelhakim [1], C.Tuleu [1], A. Coupe [2], M. Edirisinghe [1], and D. Craig [1]

[1] University College London, [2] Pfizer Ltd

2017 AAPS ANNUAL MEETING & EXPOSITION

CONTACT INFORMATION: Hend.Abdelhakim.16@ucl.ac.uk

DEVELOPING SCIENCE. IMPACTING HEALTH.

PURPOSE

Currently there is a **lack of age appropriate formulations**. Liquids can taste bad, and solids are difficult to swallow, leading to poor compliance in the paediatric population.

Manufacturing a novel dosage form that is palatable and is easy to swallow could be made possible by using a novel technique such as **electrospinning**. By combining a bitter drug, Chlorpheniramine Maleate (**CPM**), with a taste masking polymer, Eudragit E PO (**E-EPO**), taste masked fibres can be manufactured.

OBJECTIVE

Electrospinning is affected by **process**, **solution** and **environmental** factors [1]. The aim of this study is to utilise a design of experiment approach (**DoE**) to efficiently optimise the quality of the fibres produced.

METHOD(S)

All solutions were prepared to 100% using absolute ethanol. Ratios of 1:6 of CPM :E-EPO were used in the drug loaded fibres. Spraybase® electrospinning apparatus was used to manufacture the fibres. A Scanning Electron Microscope (SEM) FEI Quanta 200FEG, was used to image the fibres. ImageJ 1.46R was used to measure diameters. JMP Pro 12.0.1 was used to create the fractional design and for data analysis. Parameters investigated are shown in Table 1.

Table 1 - *A table displaying the factors and the three corresponding levels for each*

Factor	Low	Medium	High
Applied voltage (kV)	10.00	17.50	25.00
Flow rate (mL/h)	0.50	1.25	2.00
Gap distance (mm)	150.00	200.00	250.00
E-EPO concentration (%w/v)	25.00	35.00	45.00
Water in solvent (%v/v)	0.00	10.00	20.00

The desired fibres were manufactured with the morphology and ease of operation as the optimisation criteria, namely **reducing beading**, and **fibre diameter**.

RESULT(S)

Initially placebo fibres were optimised, as shown in Figs 1a, 1b and 1c.

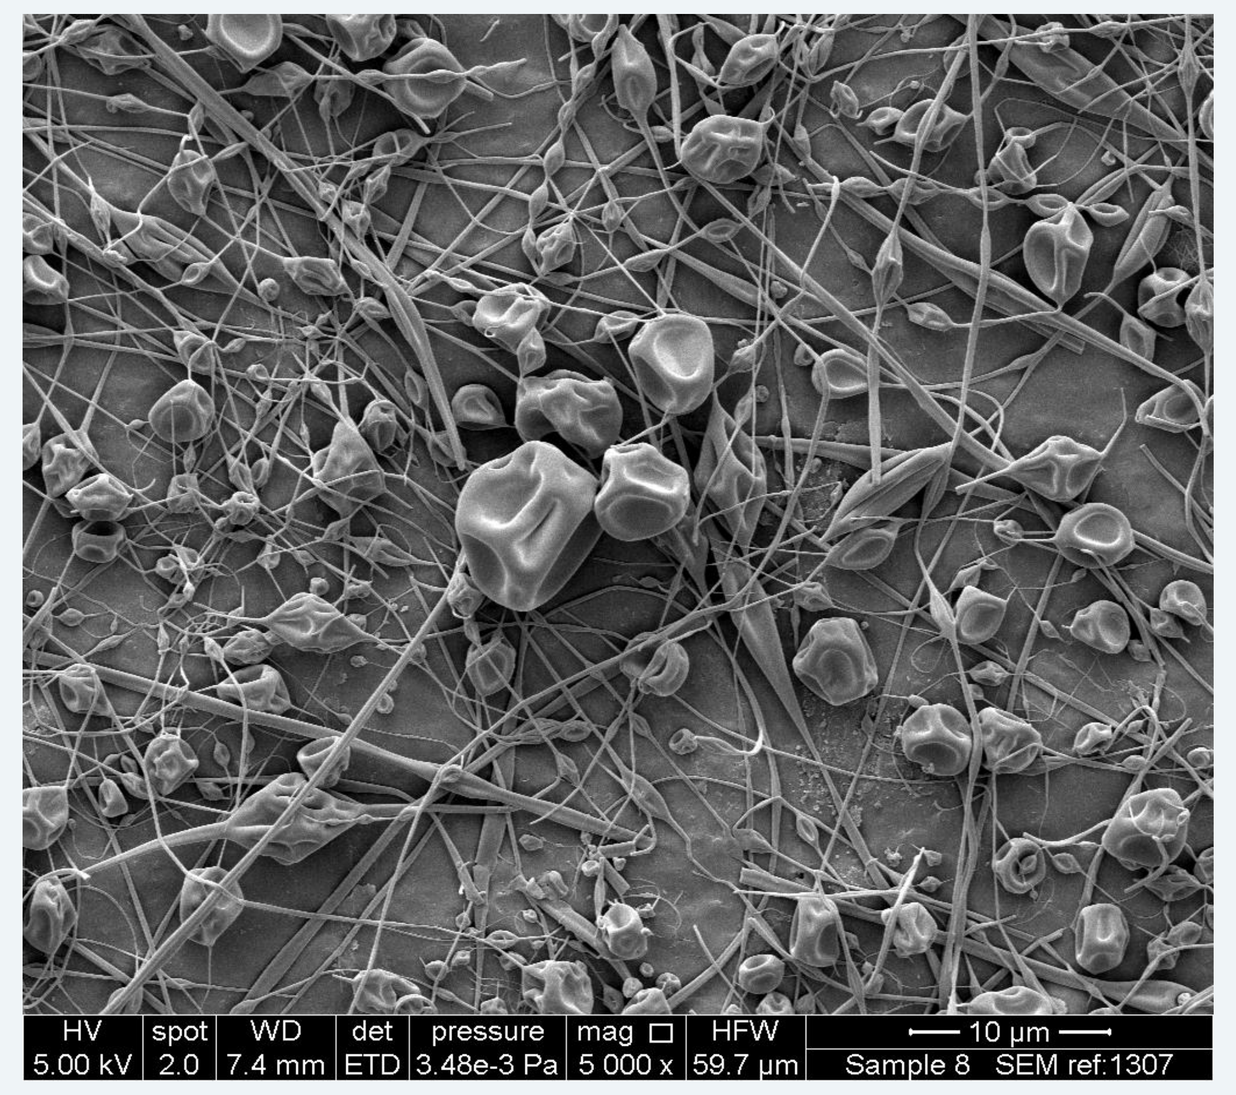

Fig. 1a - *SEM image of 25% E-EPO, in 0% Water, processed at 25 kV, gap distance of 250 mm and flow rate of 2mL/h,. Mean diameter =* ***416 ± 359 nm*** *, beading = 150/ 3 mm². Temp : 24º, RH: 27%*

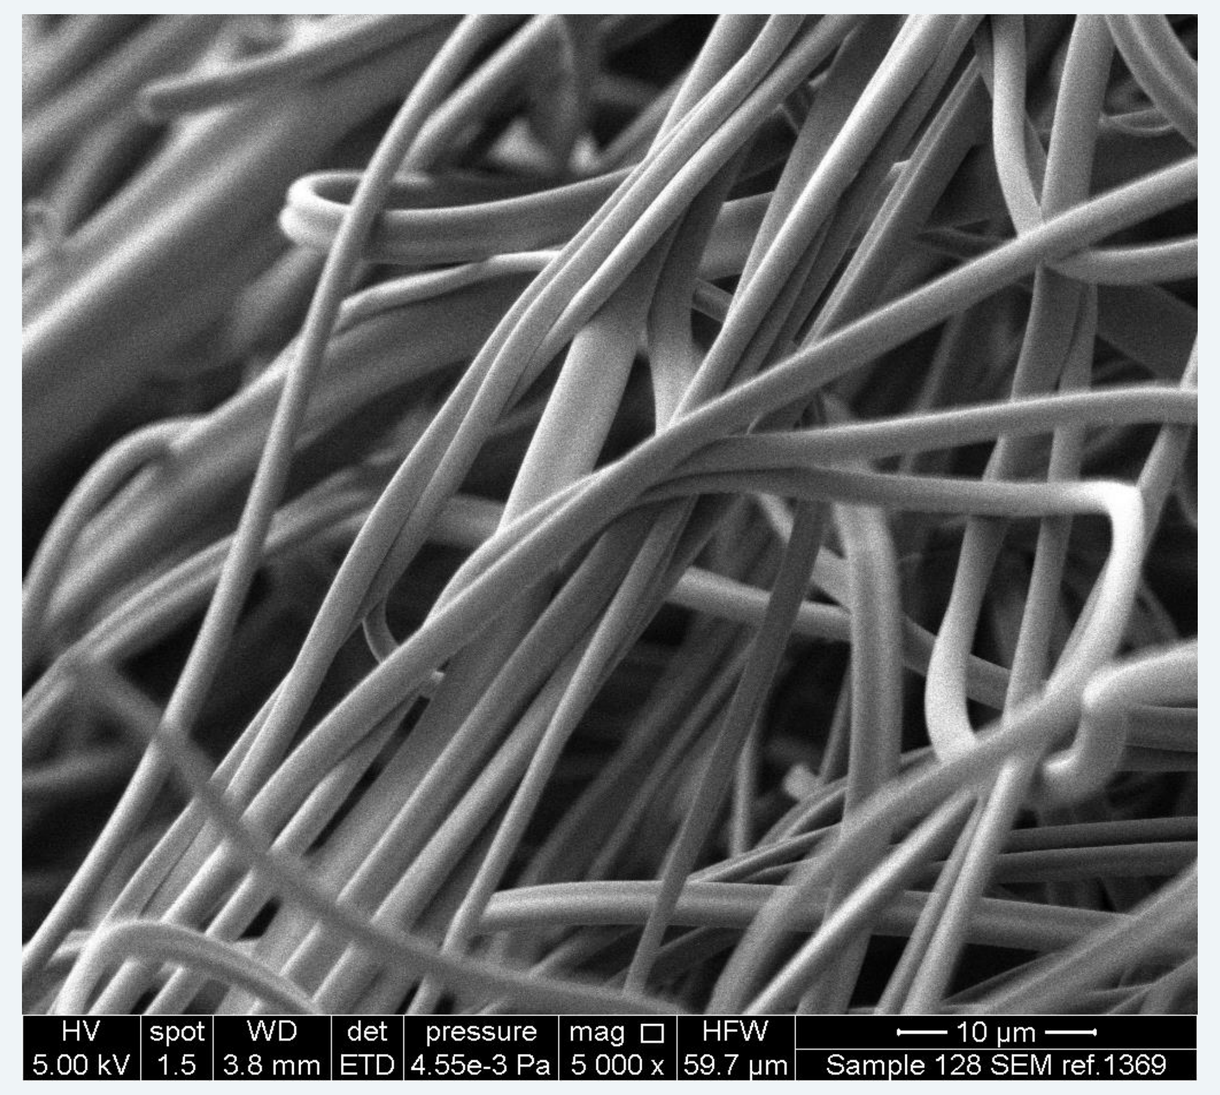

Fig. 1b - *SEM image of 35% E-EPO, in 20% Water, processed at 25 kV, gap distance of 150 mm and flow rate of 0.5mL/h. Mean diameter =* ***1508 ± 250 nm****. Temp : 28º, RH: 36%*

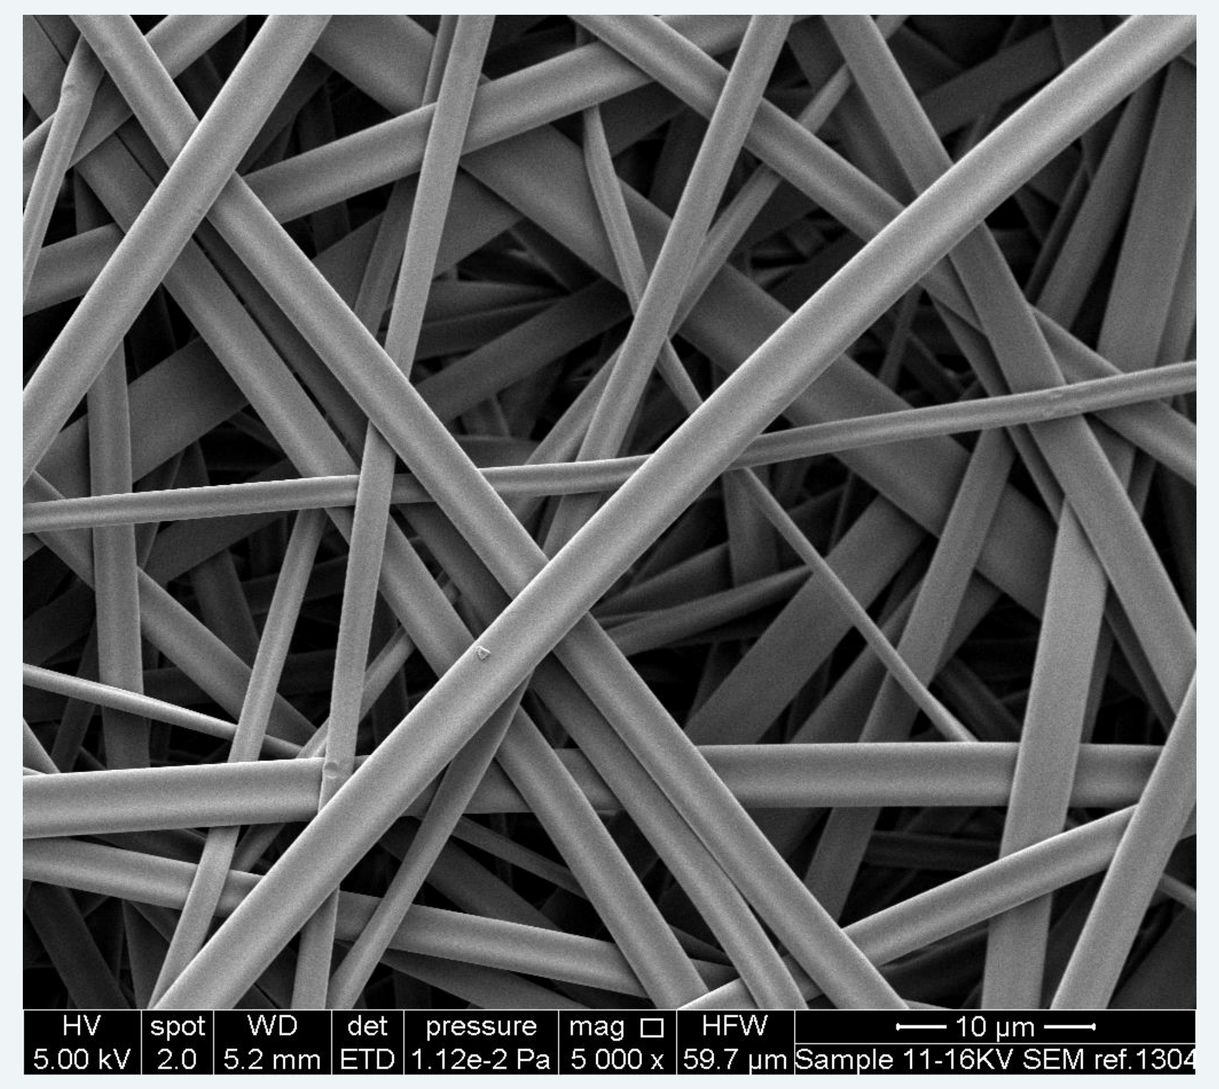

Fig. 1c - *SEM image of 45% E-EPO, in 20% Water, processed at 16 kV, gap distance of 150 mm and flow rate of 0.5mL/h, Mean diameter =* ***2789 ± 677 nm****. Temp : 24º, RH: 27%*

DoE modelling tool predicted optimum conditions that maximise desirability - **35%** E-EPO, **0.5mL/h** flow rate , **150 mm** gap distance, **15-20 kV** applied voltage were predicted to form fibres with an average diameter of 621 nm and minimal beading. When validated experimentally smooth non-beaded fibres with an average diameter of 1508 nm (± 250) were formed (Fig 1b).

When the drug was added, the solution properties changed and hence re-optimisation was required. SEM images shown in Figs 21, 2b and 2c.

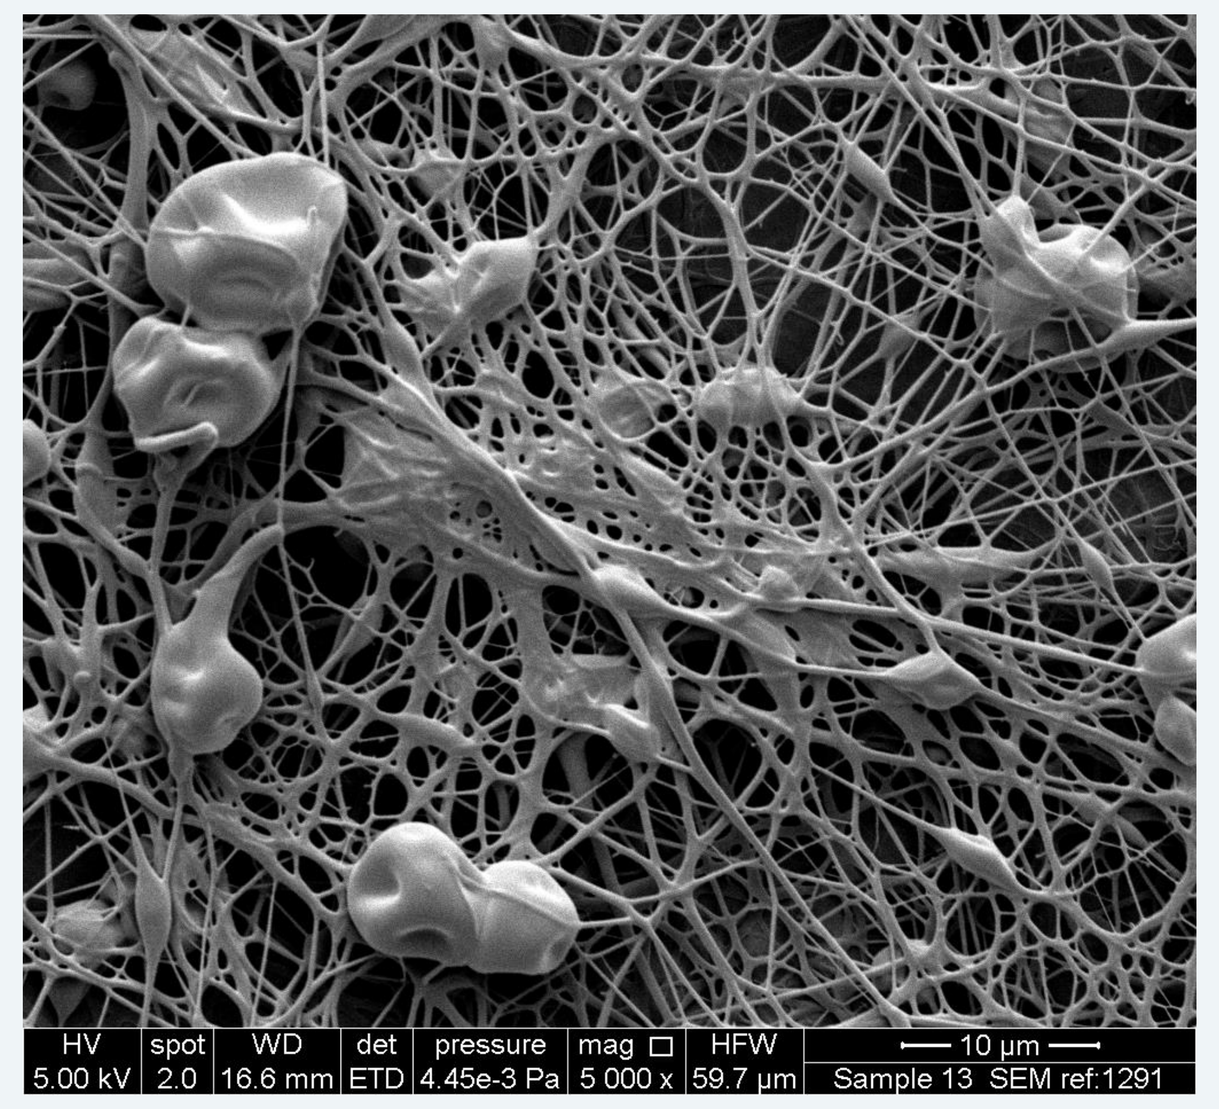

Fig. 2a – *SEM image of 25% E-EPO, in 20% Water, processed at 20 kV, gap distance of 150 mm and flow rate of 1mL/h,. Mean diameter =* ***265 ± 124 nm*** *. Temp : 24º, RH: 30%*

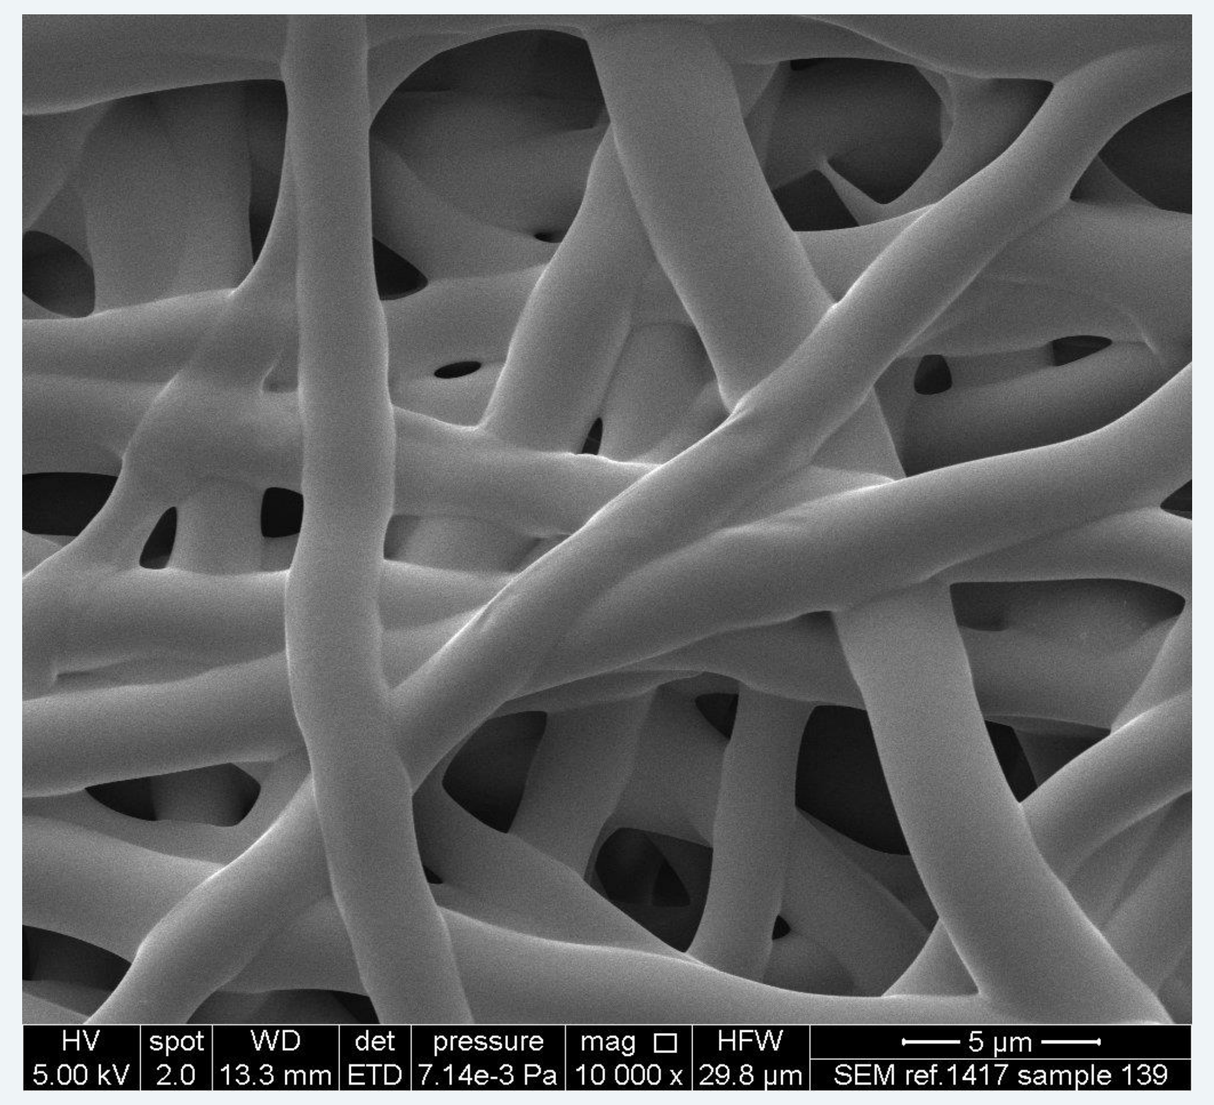

Fig. 2b – *SEM image of 35% E-EPO, in 0% Water, processed at 15 kV, gap distance of 175 mm and flow rate of 1mL/h,. Mean diameter =* ***2360 ± 326 nm****. Temp : 24º, RH: 46%*

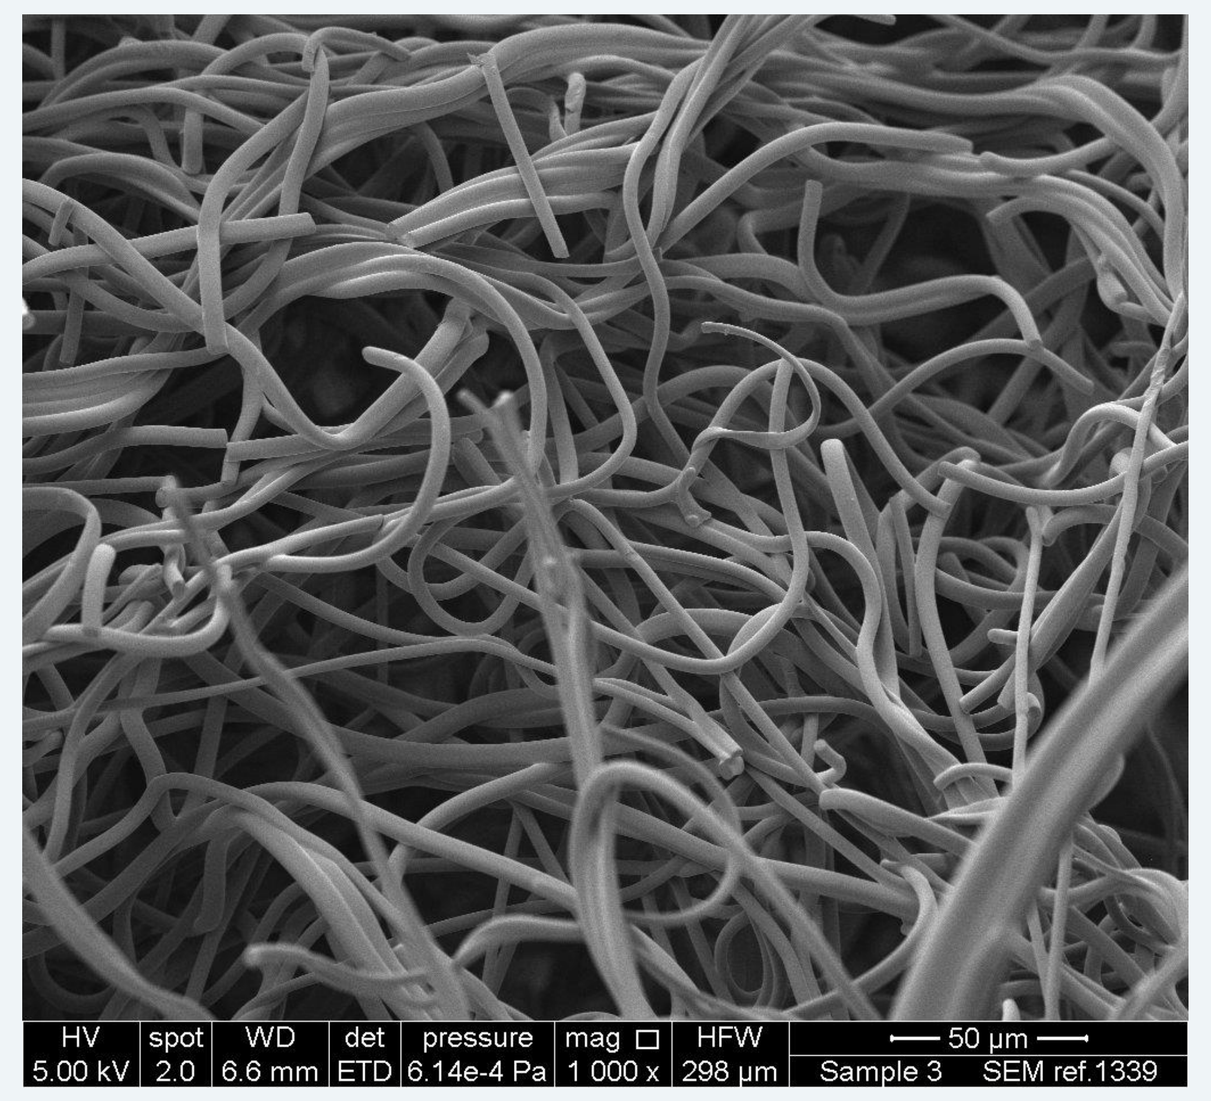

Fig. 2c – *SEM image of 40% E-EPO, in 0% Water, processed at 10 kV, gap distance of 150 mm and flow rate of 0.5mL/h,. Mean diameter =* ***3237 ± 0.667 nm****. Temp : 23º, RH: 34%*

Increased polymer concentration was found to be the most significant factor in increasing placebo fibre diameter, **p-value = 0.008**, and decreasing beading, **p-value = 0.004**.

Similar to placebo fibres, concentration was found to be the most influential factor affecting fibre morphology. At a concentration of 40% E-EPO, smooth ribbons were formed, however, needle clogging halted the smooth running of the electrospinning process

Generally, the conditions for producing good drug-loaded fibres were the same as with placebo fibres, when the drug was added at a 1:6 ratio to the polymer.

CONCLUSION(S)

The fractional factorial DoE approach reduced the number of runs required to optimise the placebo fibres from 3^5 or 243 runs, to 12 runs. The design identified the most influential manufacturing conditions for nano-diameter fibres and absence of beading as well as the prediction of optimal conditions for both outcomes. The study has therefore not only suggested parameters for producing E-EPO fibres but has also developed an approach for the rational choice of electrospinning conditions.

FUNDING

This study is funded by MRC iCASE award No. 170156, and Pfizer Ltd. Award No. 173803

REFERENCE

[1] S.R. Coles, D.K. Jacobs, M.O. James, G. Barker, A.J. Clark, K. Kirwan, J. Stanger, N. Tucker, A design of experiment approach to material properties optimization of electrospun nanofibres, J. Appl. Polym. Sci. 117 (2010) 2251–2257. doi:10.1002/app.

MRC Medical Research Council

Pfizer

UCL SCHOOL OF PHARMACY